Sphingosine 1-Phosphate Receptor Modulators as a Potential Treatment Option in COVID-19 Induced Acute Respiratory Distress Syndrome: Mechanistic Insights and Benefit-Risk Assessment

C10

Thomas Hach[1]*, Kasra Shakeri-Nejad[2]*, Marc Bigaud[2], Frank Dahlke[1], Massimiliano de Micco[3], Olivier Petricoul[2], Gordon Graham[1], Daniela Piani-Meier[1], Renato Turrini[3], Volker Brinkmann[4], Ferdinando Nicoletti[5,6]

*These authors contributed equally

[1]Novartis Pharma AG, Basel, Switzerland; [2]Novartis Institutes for Biomedical Research, Basel, Switzerland; [3]Novartis Farma S.p.A., Origgio, Italy; [4]Independent Consultant, Freiburg, Germany; [5]Department of Physiology and Pharmacology, University Sapienza of Rome, Italy; [6]IRCCS Neuromed, Pozzilli, Italy

BACKGROUND

- COVID-19, an acute respiratory disease, is caused by the newly emergent human infectious severe acute respiratory syndrome coronavirus 2 (SARS-CoV-2)[1,2]
- Globally, as of mid-May 2020, there have been approximately 4,731,000 confirmed cases of COVID-19, including 315,000 deaths[3]
- Illness severity ranges from mild to critical[4,5]
 - Asymptomatic SARS-CoV-2 infection appears to be common[4,5]
 - Symptoms often include fever, cough, and fatigue, which can intensify quickly, resulting in hospital admission[6-8]
 - ~19% of patients are hospitalized; 5% need intensive care unit treatment, with most of these patients requiring mechanical ventilation[4,5]
 - Acute respiratory distress syndrome (ARDS) in the critical cases is the leading cause of death with a high mortality rate of 40-50%[8,9]
- Mortality risk is often associated with viral-induced hyperinflammation and an accompanying cytokine release syndrome or cytokine storm (**Figure 1**)[8]
 - COVID-19 patients with ARDS typically present with total lymphocyte level significantly below the normal range, with approximately 70% of these patients showing lymphopenia (lymphocyte count, 0.8 cells/nL [interquartile range, 0.6-1.1]).[8] Low CD8+ T cells and high CD4+/CD8+ ratio are predictors of COVID-19 severity[10]
- With no approved medication numerous trials to treat or prevent COVID-19 have been initiated,[4,5] and the US Food and Drug Administration has granted Emergency Use Authorization for some existing drugs[11]
- Very recently, first case report of COVID-19 in a multiple sclerosis patient under fingolimod treatment was published. The patient revealed severe bilateral interstitial pneumonia on chest CT scan, but recovered rapidly[12]
- Efficacy of fingolimod, a non selective sphingosine 1-phosphate (S1P) receptor modulator, taken once-daily, for three consecutive days in patients with COVID-19 is currently being evaluated in a Phase 2 clinical study in China[13]

Figure 1. Pathogenesis of COVID-19 infection leading to cytokine storm in severe cases

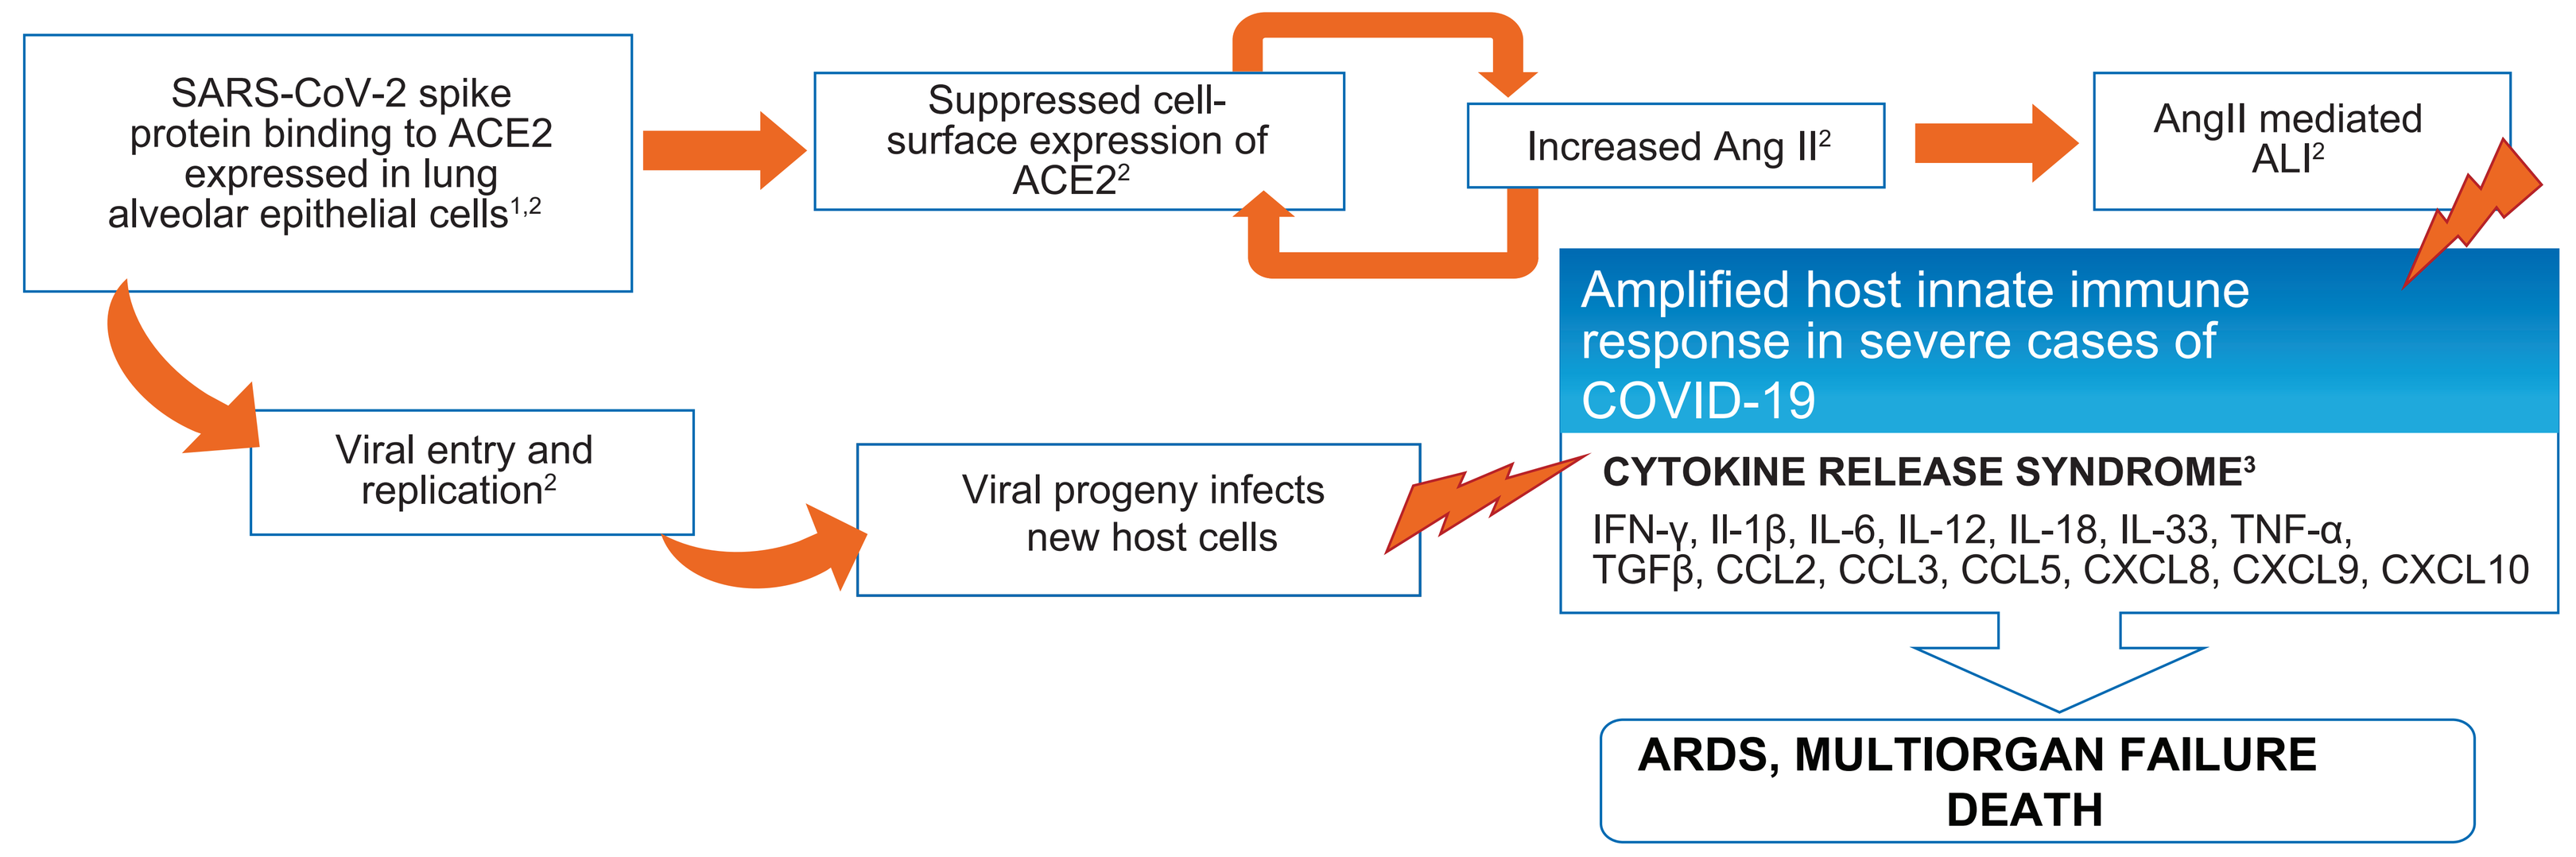

SARS-CoV-2 spike protein binding to ACE2 expressed in lung alveolar epithelial cells1,2
Suppressed cell-surface expression of ACE22
Increased Ang II2
AngII mediated ALI2
Viral entry and replication2
Viral progeny infects new host cells
Amplified host innate immune response in severe cases of COVID-19
CYTOKINE RELEASE SYNDROME3
IFN-γ, Il-1β, IL-6, IL-12, IL-18, IL-33, TNF-α, TGFβ, CCL2, CCL3, CCL5, CXCL8, CXCL9, CXCL10
ARDS, MULTIORGAN FAILURE DEATH

ACE2, angiotensin converting enzyme 2; ALI, acute lung injury; Ang II, angiotensin II; ARDS, acute respiratory distress syndrome; SARS-CoV-2, severe acute respiratory syndrome coronavirus 2

[1]Wan Y, et al. *J Virol*. 2020;94:e00127–20. [2]Kuba K, et al. *Nat Med*. 2005;11:875–879. [3]Sarzi-Puttini P, et al. *Clin Exp Rheumatol*. 2020;38(2):337–342.

OBJECTIVES

- To review the potential mechanisms by which fingolimod may regulate the inflammatory response to SARS-CoV-2
- To assess the potential benefit-risk of short-term treatment (3-days, once daily) with fingolimod 0.5 mg in patients with COVID-19 experiencing ARDS

METHODS

- Review of the existing evidence of S1P receptor modulators regarding potential benefits for patients with COVID-19
- Pharmacokinetic-Pharmacodynamic (PKPD) simulation of lymphocyte count with short-term fingolimod treatment
 - A baseline lymphocyte count of 0.7 cells/nL was used for these simulations. Dosing regimens with once daily (q.d.) fingolimod 0.5 mg for 3, 4 and 5 days were simulated. PK profiles and lymphocytes effects are displayed for 3-days dosing

Role of S1P in viral-induced hyperinflammation

- Sphingolipids assume multiple roles in the regulation of neutrophil chemotaxis, neutrophil apoptosis, as well as endothelial and epithelial barrier functions in ARDS (**Table 1**)[14]
- S1P has two effects on neutrophils: an early attenuation of chemotaxis and later survival promotion[14,15]
- S1P also mediates bidirectional regulation of vascular permeability via its G-protein-coupled receptors[14]
 - The barrier function is enhanced by S1P1 but impaired by S1P2 and S1P3 receptor

Table 1. Role of S1P in hyperinflammation during acute phase of ARDS

Mediator	Neutrophil chemotaxis	Endothelial permeability	Neutrophil Apoptosis	Epithelial permeability
NSMase*				
ASMase*	↑		↓	
S1P		↑		↓
S1P1R	↓		↓	
S1P2R		↓		↓
S1P3R				↑
S1P4R		Unknown		↑

*catalyses the breakdown of sphingomyelin to ceramide and phosphorylcholine

ARDS, acute respiratory distress syndrome; ASMase, acid sphingomyelinase; NSMase, neutral sphingomyelinase; S1P, sphingosine-1-phosphate; S1P1-4R, type 1-4 S1P receptors

Fingolimod: Potential mechanism of action in COVID-19

- Therapies targeting S1P-receptors act pharmacologically as agonists on the receptors. Fingolimod is a S1P1, 3, 4, and 5 receptor agonist but, as it down-modulates the S1P1 receptor, it is functionally a S1P1 antagonist[15,16]
- Short-term intervention with fingolimod can attenuate maladjusted immune responses against SARS-CoV-2 via following mechanisms (**Table 2**)
 - **S1P1-mediated regulation of lymphocyte trafficking:** Down-modulation (through internalisation) of S1P1 receptors expressed on the lymphocytes results in rapid reduction in absolute lymphocyte count (ALC) and reduced infiltration of cytotoxic T cells, including inflammatory Th17 lymphocytes
 - **S1P2 and S1P3-dependent increase in endothelial and epithelial permeability:** The equilibrium between S1P1, S1P2 and S1P3 tones at the level of endothelial and epithelial barriers is displaced towards increased S1P1 tone and reduced barrier permeability
- In preclinical models, fingolimod attenuated cytokine release via pathways independent of S1P receptor modulation (**Table 2**)
 - **Activation of serine/threonine protein phosphatase 2A (PP2A):** Mitogen-activated protein kinase (MAPK) signaling pathways are involved in cytokine production. PP2A regulates the MAPK pathways. Fingolimod increases basal PP2A phosphatase activity and significantly represses IL-6 and IL-8 mRNA expression (**Figure 2**) and downstream CXCL1 and CXCL2 secretion
 - **Increased c-Abl kinase activity**: Fingolimod enhances pulmonary vascular endothelial barrier function by significantly increasing c-Abl kinase activity; this pathway does not require adherens junction or tight junction protein complexes (**Figure 3**)

Table 2. Potential mechanisms of short-term exposure of S1PR modulators in COVID-19 ARDS

	$S1P_1$ receptor - modulated effects		Non S1P receptor - modulated effects	
Chemotaxis/ immune response	**Reduction in Th-17 lymphocytes**	Reduced tissue infiltration release of IL-17 and downstream pro-inflammatory cytokines and chemokines[1,2]	**Increased PP2A[6,7]**	Suppresses IL-6 and IL-8 cytokine secretion in human alveolar epithelial cell lines
	Enrichment of of T_{regs} and B_{regs} (via sparing of this subpopulation)	Shift towards anti inflammatory response[3,4]		Decreases downstream CXCL1 and CXCL2 release
	Innate immune cells	Reduced pro-inflammatory cytokines[1]		
Endothelial permeability	**Modulation of the endothelial barriers**	Barrier enhancement[5]	**c-Abl tyrosine kinase[8]**	Barrier enhancement by increased transendothelial electrical resistance
Potential therapies	Fingolimod, siponimod, ozanimod, ponesimod		Fingolimod	

IL, interleukin; PP2A, protein phosphatase 2A; S1P, sphingosine-1-phosphate; Th, helper T cell; T_{regs}, regulatory T cell; B_{regs}, regulatory B cell; SEM, standard error of mean

[1]Teijaro JR, et al. *Cell* 2011;146:980–991. [2]Mehling M, et al. *Neurology* 2010;75:403–410. [3]Grützke B, et al. *Ann Clin Transl Neurol*. 2015;2(2):119–130. [4]Wu Q, et al. *JCI Insight*. 2020;5:e134251. [5]Brinkmann, V, et al. *Am J Transplant* 2004;4: 1019–1025. [6]Rahman MM, et al. *Sci Rep*. 2016;6:37297. [7]Rahman MM, et al *Sci Rep* 2015;5:10063. [8]Wang L, et al. *Eur Respir* J. 2011;38:78–88.

Figure 2. Suppressed cytokine secretion by fingolimod via PP2A activation

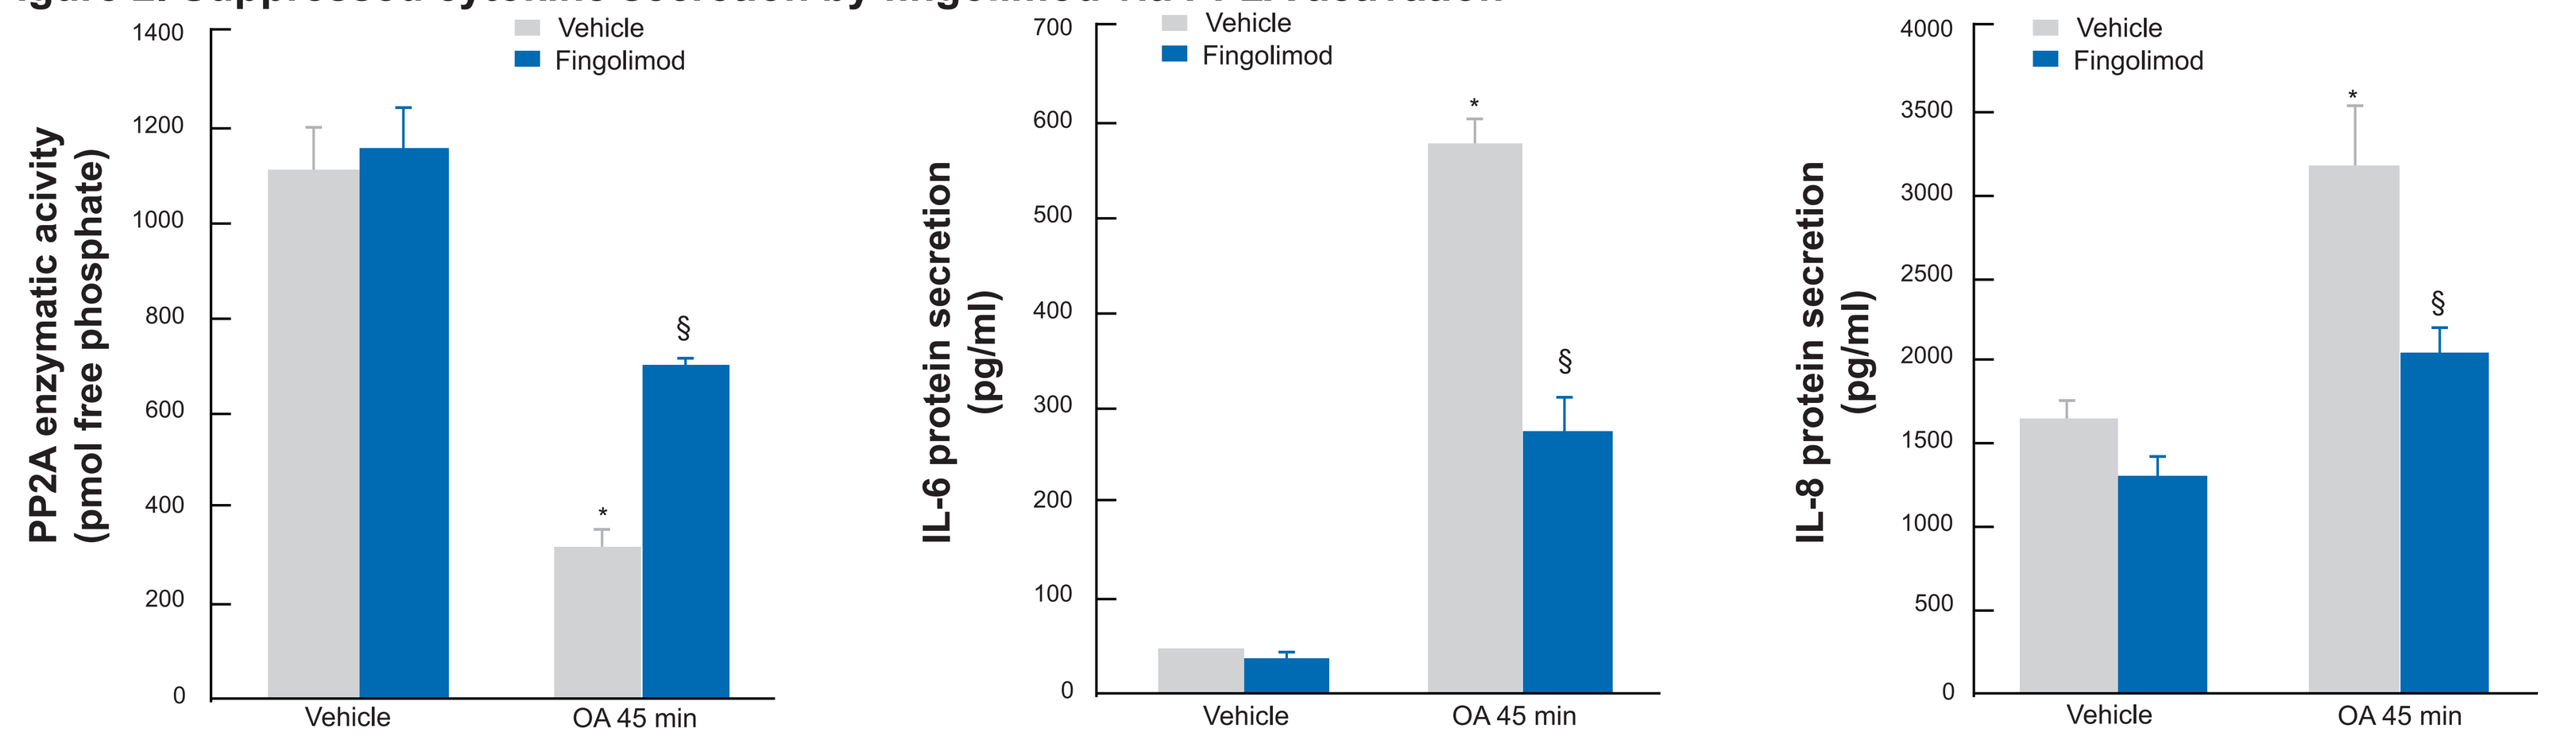

Vehicle
Fingolimod
PP2A enzymatic acivity (pmol free phosphate)
IL-6 protein secretion (pg/ml)
IL-8 protein secretion (pg/ml)
Vehicle
OA 45 min

IL, interleukin; OA, okadaic acid; PP2A, protein phosphatase 2A
A549 cells were treated for 6 h with 2.5 µM fingolimod prior to 45 min with 1 µM OA, compared to vehicle.
* denotes a significant effect of OA or § fingolimod; p<0.05. Data are mean+SEM values from three independent experiments.
Figure adapted from Rahman MM, et al. *Sci Rep* 2015;5:10063.

Figure 3. Barrier enhancement by fingolimod via c-Abl tyrosine kinase

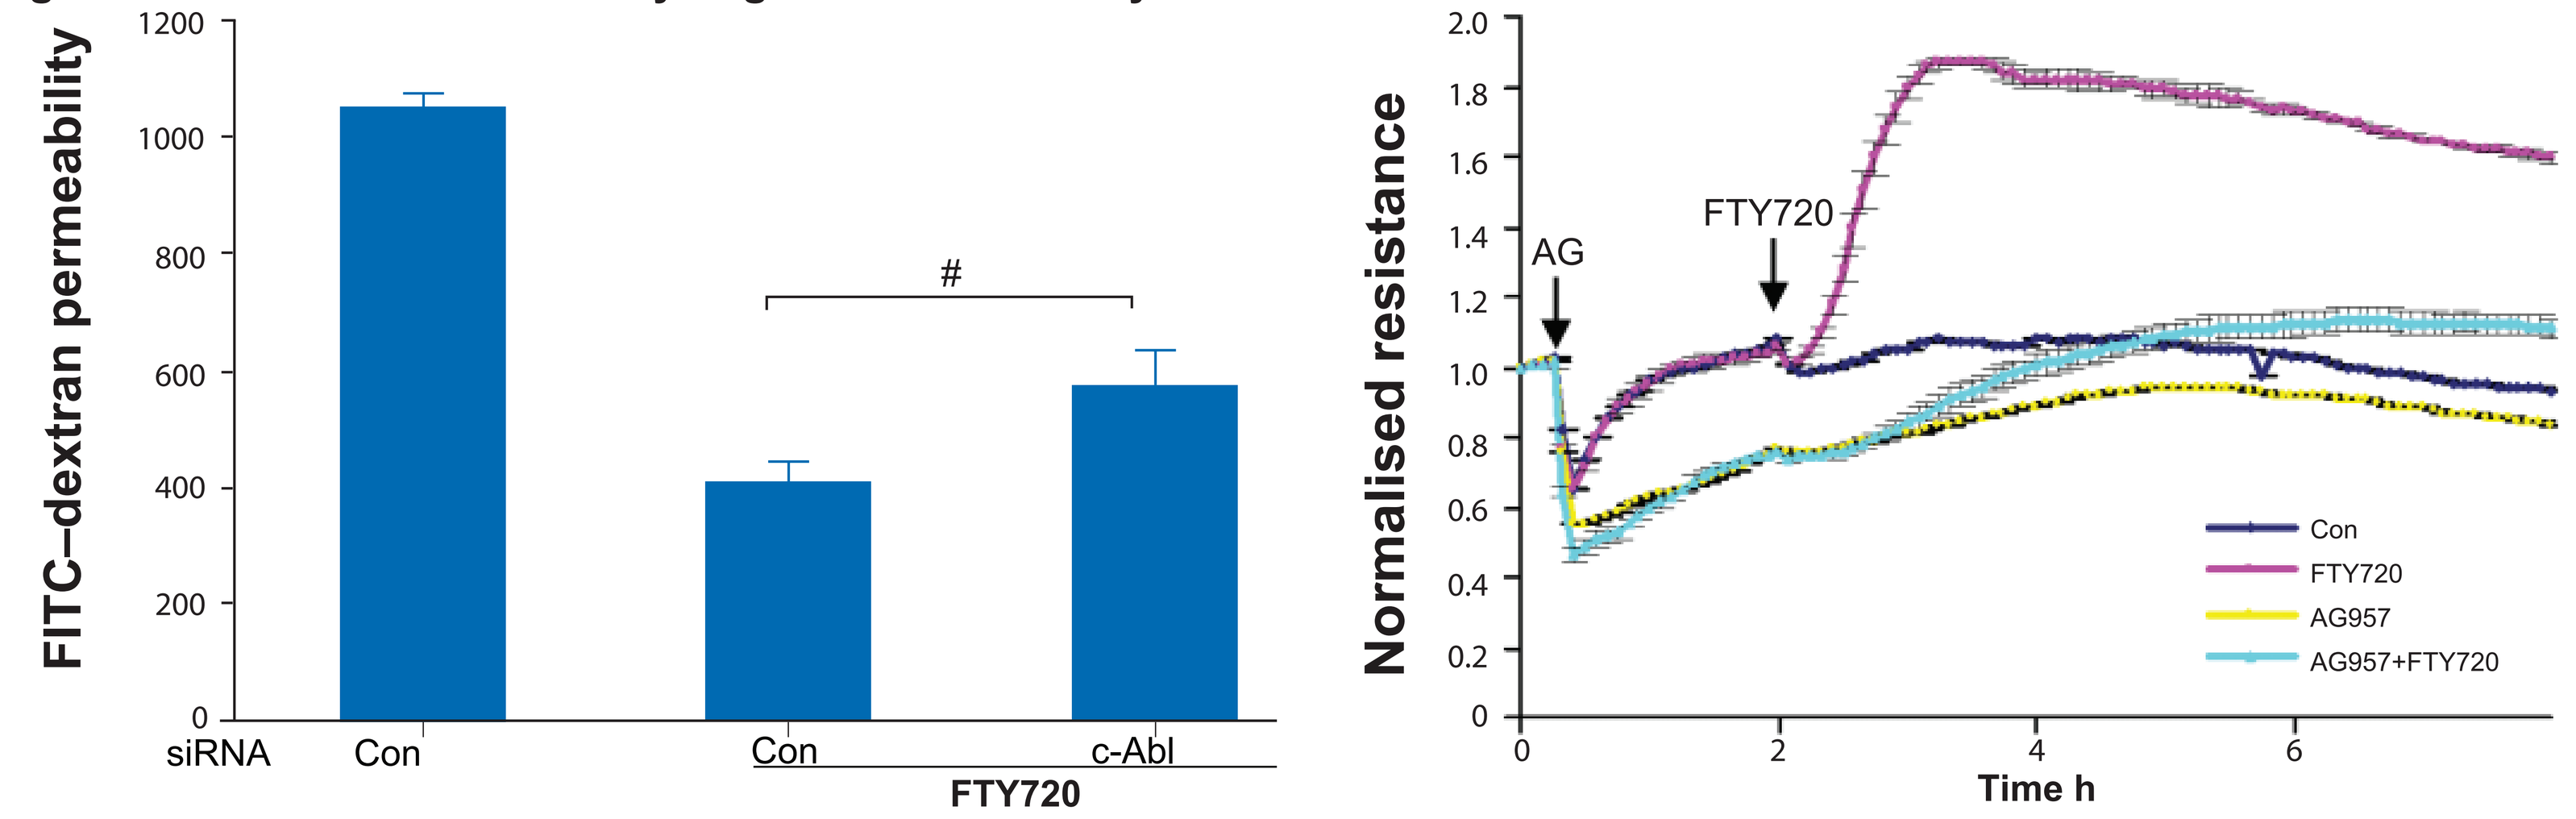

FITC–dextran permeability
siRNA
Con
Con
c-Abl
FTY720
Normalised resistance
AG
FTY720
Time h
Con
FTY720
AG957
AG957+FTY720

Con, control; FITC, fluorescein isothiocyanate; SEM, standard error of mean; FTY720, fingolimod; AG957 is a c-Abl inhibitor
Human pulmonary artery endothelial cells were stimulated with fingolimod (1 µM). *p<0.031 versus Con siRNA. Data are mean+SEM values from three independent experiments.
Figure adapted from Wang L, et al. *Eur Respir* J. 2011;38:78–88.

PKPD simulation of lymphocyte count with short-term fingolimod dosing

- Fingolimod q.d. dosing for 3 days is expected to reduce circulating lymphocyte levels from 0.7 to nadir of 0.29 cells/nL, already reaching levels of 0.37 cells/nL within 6-8 h after the first dose (**Figure 4A**)
- Lymphocyte count returns to levels above 0.5 cells/nL within 10 days after last dose and to baseline values by 40-60 days (**Figure 4B**)
- Dosing over 4 or 5 days only slightly lowers the nadir (0.27 and 0.26 cells/nL, respectively), but lymphocyte recovery to 0.5 cells/nL is prolonged to approximately 15 and 18 days, respectively. Hence the proposed 3-days dosing regimen was considered optimal for this acute treatment setting

Figure 4. Fingolimod 0.5 mg q.d. for 3 days

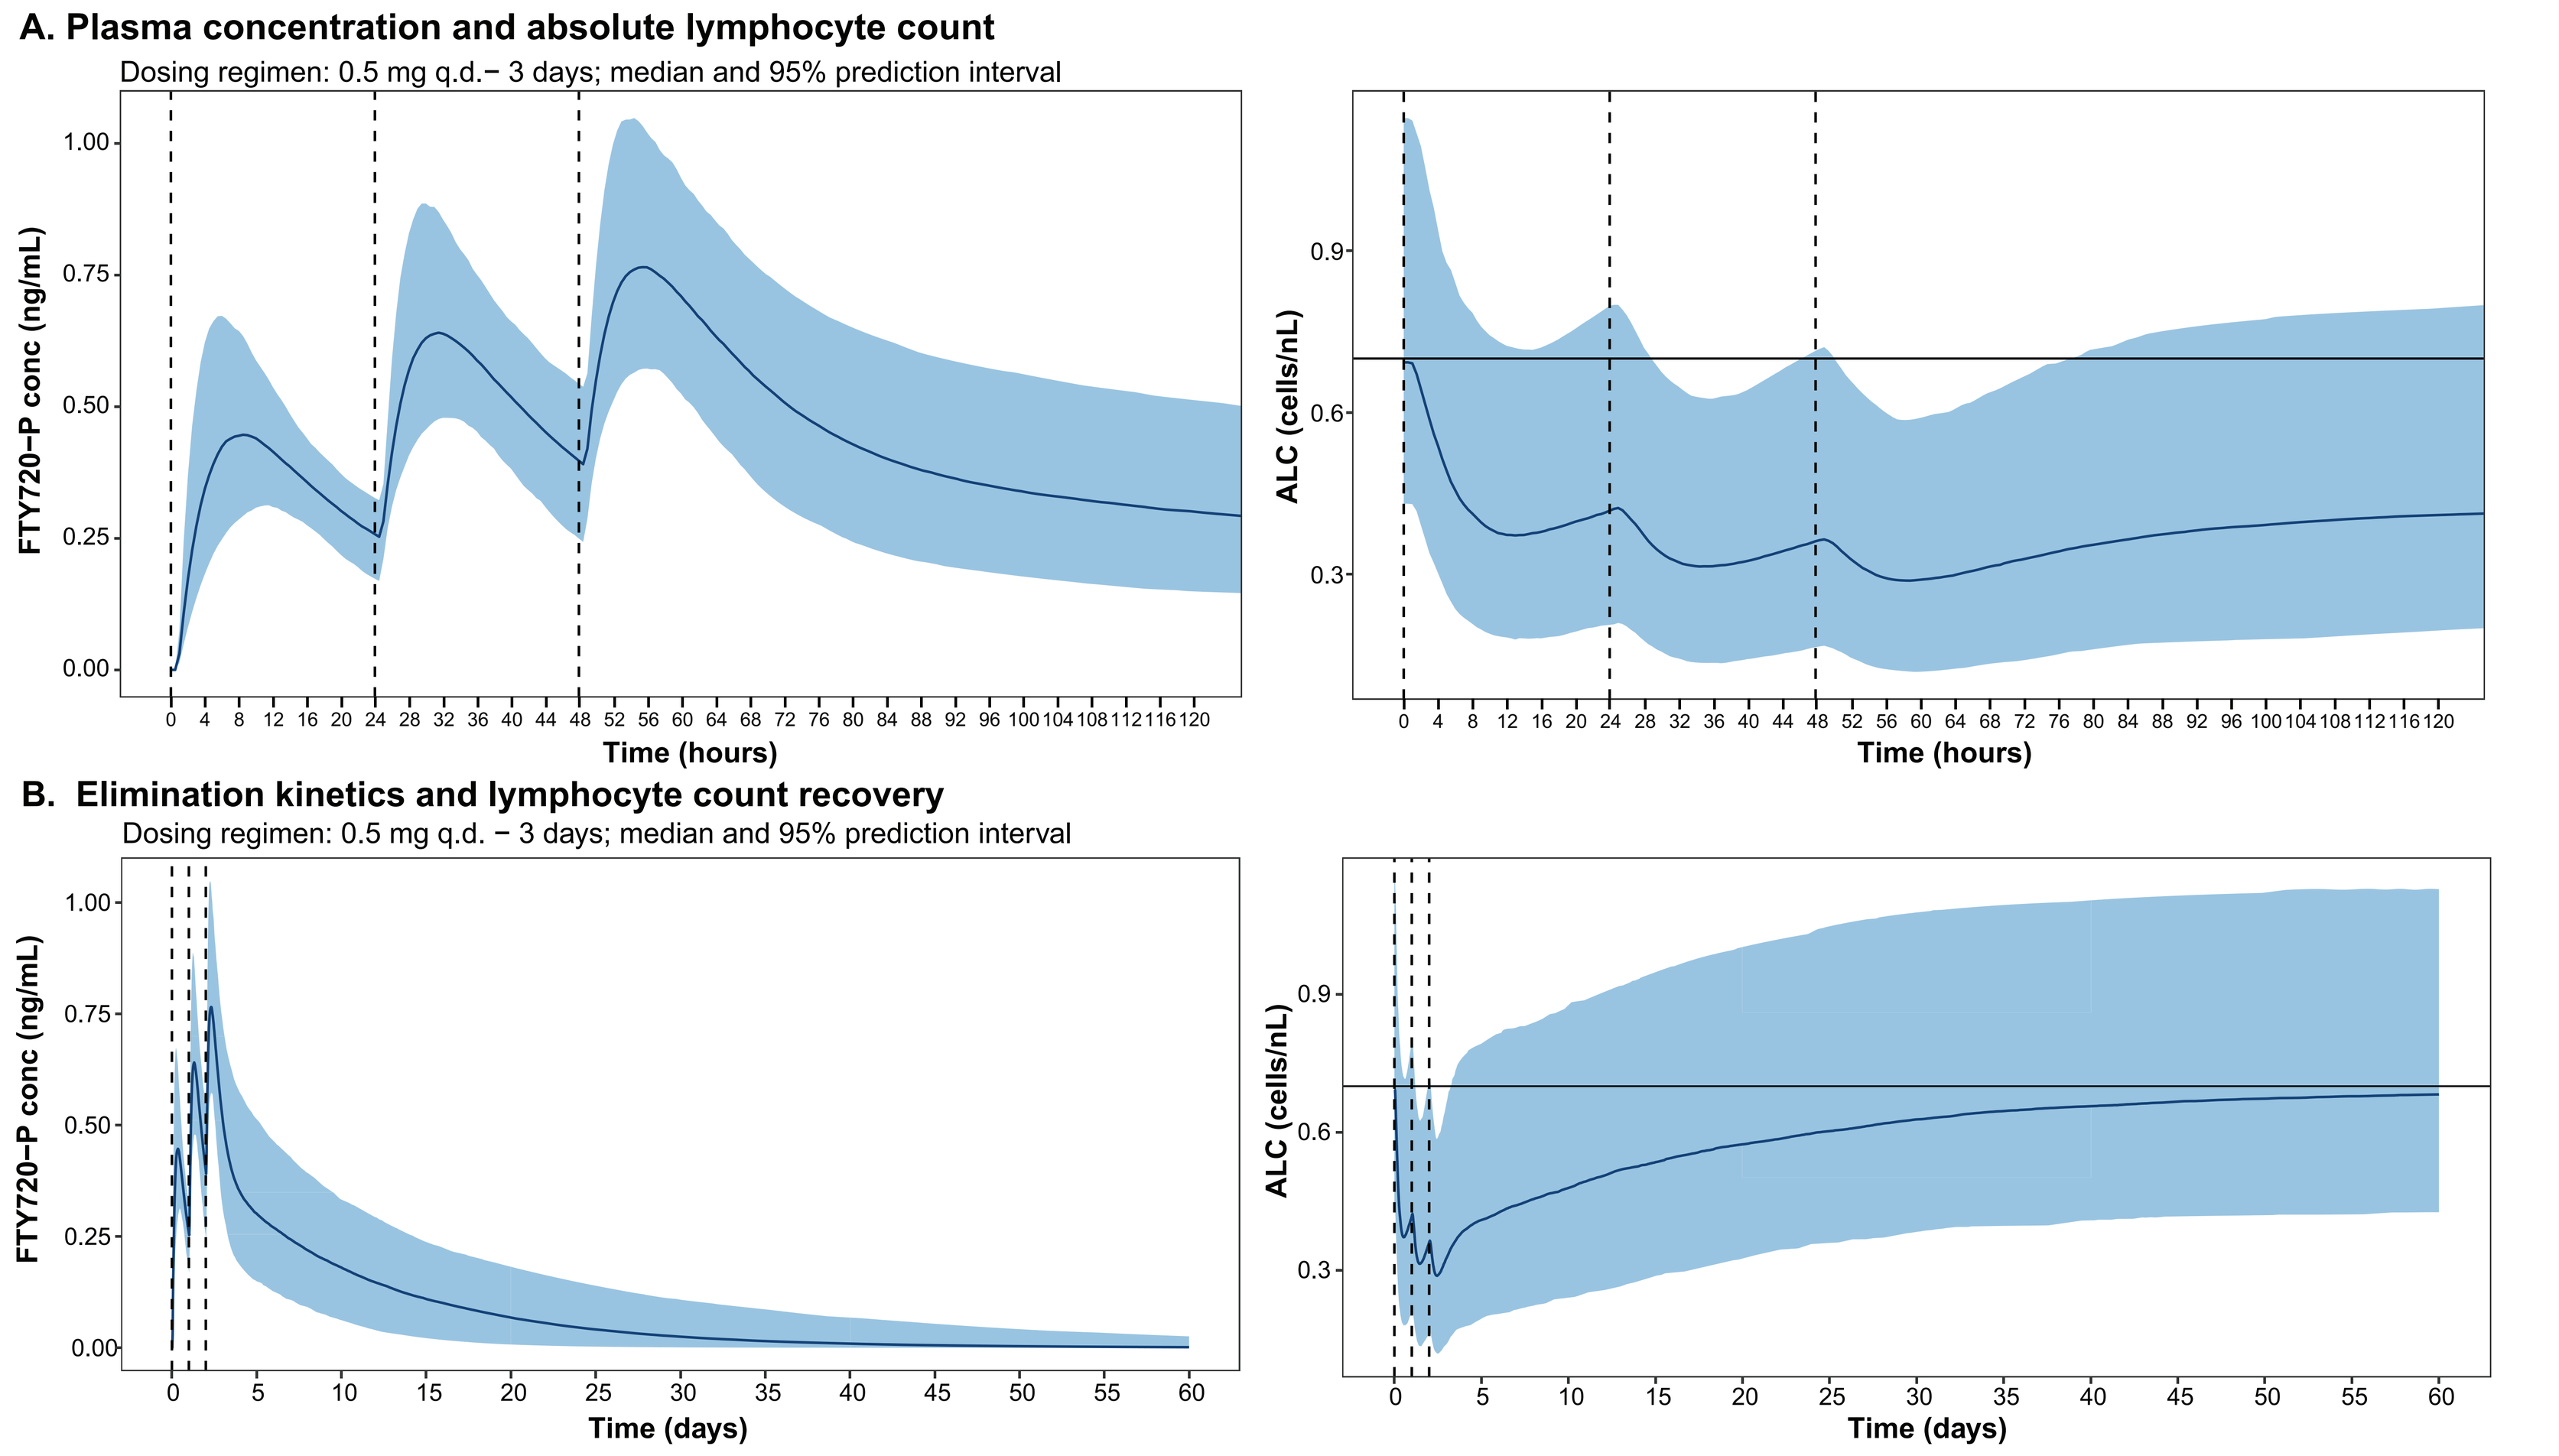

A. Plasma concentration and absolute lymphocyte count
Dosing regimen: 0.5 mg q.d.– 3 days; median and 95% prediction interval
FTY720–P conc (ng/mL)
ALC (cells/nL)
Time (hours)
B. Elimination kinetics and lymphocyte count recovery
Dosing regimen: 0.5 mg q.d. – 3 days; median and 95% prediction interval
FTY720–P conc (ng/mL)
ALC (cells/nL)
Time (days)

ALC, absolute lymphocyte count; FTY720-P, fingolimod-phosphate; q.d., once daily

CONCLUSIONS

- Both S1P receptor-mediated and non S1P receptor-mediated mechanisms are involved in the regulation of the host-inflammatory response to SARS-CoV-2
- A short-term intervention with fingolimod might rapidly attenuate maladjusted immune responses
 - The sequestration of cytotoxic lymphocytes along with strengthening of the endothelial barrier potentially reduces chemotaxis and immune response in the acute phase of ARDS
- Sparing of effector memory immune responses may relate to relatively low risk of infections
- The known effects on heart rate and cardiac rhythm by fingolimod could be managed under the intensive care setting for ARDS management in these patients
- Fingolimod 0.5 mg q.d. for 3 days results in an early onset of pharmacological effects within the first few hours which are maintained for up to 10 days and the treatment effects may be considered sufficient for the acute setting of ARDS patients
- Fingolimod and other S1P receptor modulators, such as siponimod, may represent a potential treatment option to ameliorate immune responses against SARS-CoV-2; however, further studies in these critically ill patients and careful benefit-risk evaluation is warranted

References

1. Guo YR, et al. *Mil Med Res*. 2020;7(1):11.
2. Jin Y, et al. *Viruses*. 2020;12(4).
3. COVID-19 Dashboard by the Center for Systems Science and Engineering (CSSE) at Johns Hopkins University (JHU) https://coronavirus.jhu.edu/map.html (Accessed on 15 May 2020).
4. European Centre for Disease Prevention and Control. https://www.ecdc.europa.eu/en/current-risk-assessment-novel-coronavirus-situation (Accessed 15 May 2020).
5. Centers for Disease Control and Prevention. https://www.cdc.gov/coronavirus/2019-ncov/hcp/clinical-guidance-management-patients.html (Accessed 15 May 2020).
6. Huang C, et al. *Lancet*. 2020;395(10223):497–506.
7. Mehta P, et al. *Lancet*. 2020;395(10229):1033–1034.
8. Wang D, et al. *JAMA*. 2020;323(11):1061–1069.
9. Ruan Q, et al. *Intensive Care Med*. 2020;46(5):846–848.
10. Wang F, et al. J. Infect. Dis. 2020;221:1762–1769.
11. US Food and Drug Administration. www.fda.gov/emergency-preparedness-and-response/mcm-legal-regulatory-and-policy-framework/emergency-use-authorization (Accessed 15 May 2020).
12. Foerch et al. MSRD Volume 42, July 2020, 102180.
13. https://clinicaltrials.gov/ct2/show/NCT04280588
14. Stefan U and Yang Y Sphingolipids in acute lung injury 2013, Volume 216.
15. Swan DJ, Kirby JA, Ali S. J *Cell Mol Med*. 2010;14(9):2211–2222.
16. Hla T, Venkataraman K, Michaud J. *Biochim Biophys Acta*. 2008;1781(9):477–482.

Disclosures

The study was supported by Novartis Pharma AG, Switzerland. Volker Brinkmann and Ferdinando Nicoletti have nothing to disclose. Thomas Hach, Kasra Shakeri-Nejad, Marc Bigaud, Frank Dahlke, Massimiliano de Micco, Olivier Petricoul, Gordon Graham, Daniela Piani-Meier, and Renato Turrini are employees of Novartis.

Acknowledgments

The authors acknowledge COVID-19 & MS: Global Data Sharing Initiative; for details please visit https://www.msif.org/covid-19-ms-global-data-sharing-initiative/
Medical writing support was provided by Uma Kundu, Anuja Shah and design support by Ravi Kisor Babu. The final responsibility for content lies with the authors.
Poster presented at the 6th Congress of the European Academy of Neurology, May 23–26, 2020.

Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=6f977
Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address: thomas.hach@novartis.com

Text: Q6f977
To: 8NOVA (86682) US Only

+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe & Russia
+46737494608 Sweden, Europe

Scan this QR code to download a copy Poster